92

A Conceptual Model for Assessing Psychosocial Functioning in People Living With Schizophrenia

Stella Karantzoulis,[1] Julia Price,[1] Brennah Fallon,[1] Anna Krasnow,[2] Amy Claxton,[3*] Sarah Donelson,[3] Kristin Gillard,[3] Chad Gwaltney[4]

[1]IQVIA, New York, NY, US; [2]IQVIA, London, UK; [3]Bristol Myers Squibb, Princeton, NJ, US; [4]Gwaltney Consulting, RI, US

*At the time the analysis was conducted

Background

- Schizophrenia is a serious mental health disorder with symptoms and behaviors impacting all aspects of life, including hallucinations, delusions, and social withdrawal[1]
- Between 10–60% of people living with schizophrenia (PLS) are reported to have a partial response to treatment with antipsychotic drugs[2]
- As part of ongoing treatment management for PLS, clinician-reported outcomes (ClinRO) play a vital role in the assessment of symptoms to gauge treatment response when insight from PLS may be limited[3]
- This qualitative research study was conducted to:
 - Identify the published evidence describing the experience (signs, symptoms, impacts) of adults living with schizophrenia
 - Understand how clinicians describe the impacts of schizophrenia on people's daily life
 - Develop and refine a conceptual model to be used as the basis of a ClinRO assessment of psychosocial functioning in PLS, and to provide a visual representation of the impact of symptoms on daily life

Methods

Development of the conceptual model

- A targeted literature review (TLR) focused on peer-reviewed publications from 2014 to 2024 was conducted using Embase according to Preferred Reporting Items for Systematic Reviews and Meta Analyses (PRISMA) guidelines
- Studies involving adults with schizophrenia, regardless of symptom control, treatment status, or comparator use, including interventional, observational, qualitative studies, and reviews were included
- Search terms included the signs, symptoms, and impacts of living with schizophrenia, as well as patient experience and health-related quality of life
- A dedicated search was also conducted to identify literature related to experience of PLS who had an inadequate response to antipsychotic treatment
- Concepts were grouped thematically using an iterative process, in alignment with:
 - The DSM-5 classification of symptoms[4]
 - Input from expert clinicians
 - A board-certified clinical neuropsychologist
- Insights gathered from the 2022 FDA Voice of the Patient (VoP) report[5] and the TLR were used to describe the PLS experience and inform the development of the preliminary conceptual model

Refinement of the conceptual model

- One-on-one, 60-minute, semi-structured, qualitative interviews were conducted with US-based board-certified Psychiatrists
- Clinicians required a minimum of 5 years' experience treating PLS with an active case load of > 20 PLS per month at the time of the interview
- Feedback was gathered from clinicians on:
 - The impacts of schizophrenia on people's daily life
 - The relevance and completeness of signs, symptoms, and impacts in the preliminary conceptual model
 - The clarity and appropriateness of language used (including domain and concept labels)
 - The categorization of concepts as signs/symptoms or impacts
- Interviews were recorded, transcribed, and anonymized
- Modifications were made to the preliminary conceptual model based on review of the clinician interview findings and discussion with expert clinicians

Results

TLR

- In addition to the VoP report, 10 relevant articles were identified from an initial identification of 2,463 titles; 5 articles were qualitative studies, and 5 articles used close-ended questionnaires (**Figure 1**)
- A total of 70 signs and symptoms across 7 concept domains, including positive, negative, cognitive, mood-related, and "other" behavioral symptoms, as well as harmful behaviors, and treatment-related symptoms were identified
- A total of 23 impacts across 7 concept domains were mapped
- Severity ratings of symptoms were not reported in any of the 11 sources reviewed

Figure 1. Concept literature search PRISMA diagram

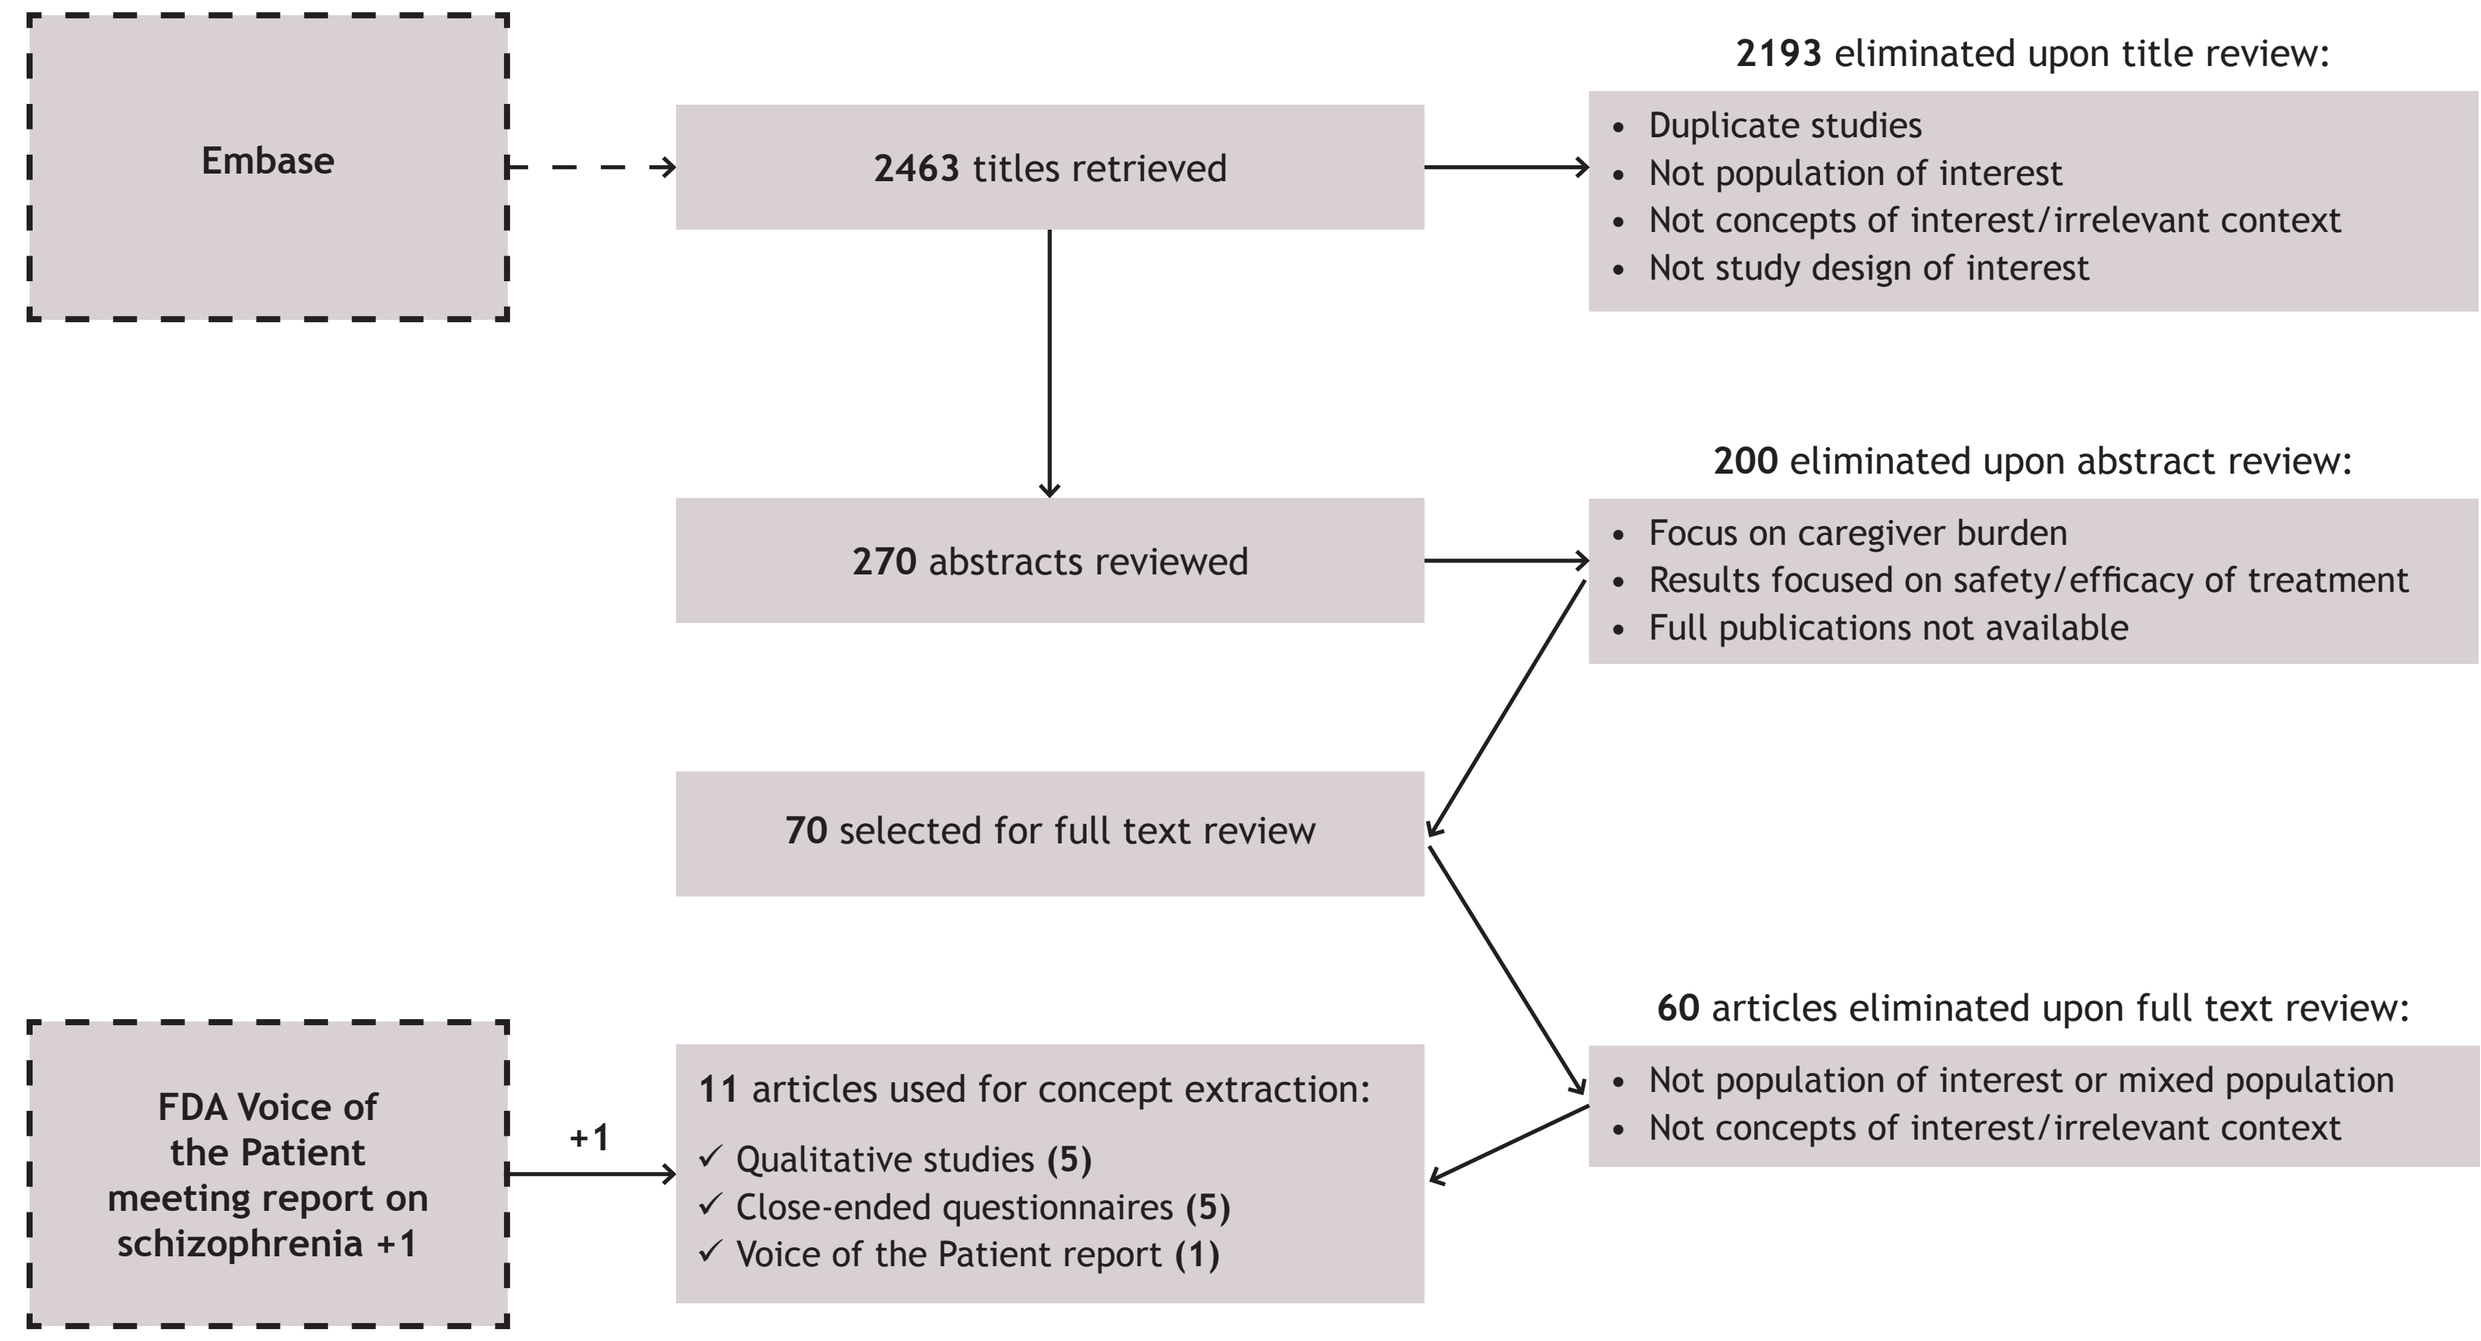
Embase
2463 titles retrieved
2193 eliminated upon title review:
• Duplicate studies
• Not population of interest
• Not concepts of interest/irrelevant context
• Not study design of interest
270 abstracts reviewed
200 eliminated upon abstract review:
• Focus on caregiver burden
• Results focused on safety/efficacy of treatment
• Full publications not available
70 selected for full text review
60 articles eliminated upon full text review:
• Not population of interest or mixed population
• Not concepts of interest/irrelevant context
FDA Voice of the Patient meeting report on schizophrenia +1
+1
11 articles used for concept extraction:
✓ Qualitative studies (5)
✓ Close-ended questionnaires (5)
✓ Voice of the Patient report (1)

Preliminary development of the conceptual model

- All concepts identified from the TLR and the VoP report were included in the preliminary conceptual model
- Signs and symptoms were categorized into 2 groups:
 - Disease-related symptoms were grouped into 6 concept categories: positive, negative, cognitive, mood-related, and "other" symptoms plus harmful behaviors
 - Cognitive symptoms were organized based on the DSM-5 classification of cognition[5]
- Overlapping treatment-related and disease-related symptoms were clearly marked
- Impacts of both disease- and treatment-related symptoms were grouped into 7 categories: emotional, day-to-day functioning, psychosocial functioning, work or school performance, relationships, sleep, and "other" impacts

Clinician perspectives on the impact of schizophrenia on PLS

- In total, 10 psychiatric clinicians were interviewed, 8 of whom practiced in both inpatient and outpatient settings, 9 had > 10 years of experience, with 4 of these having > 20 years of experience treating PLS (**Table 1**)
- Clinicians described schizophrenia as being highly burdensome to PLS, affecting nearly all aspects of life, including social, personal, and occupational functioning
- They reported that 20–70% of PLS have an inadequate treatment response to antipsychotic medication, and noted that there was a level of subjectivity around the definition of "inadequate treatment response" vs "treatment resistance"
- Symptoms described as most impactful to patients (hallucinations, delusions, neglect of hygiene, social withdrawal, cognitive deficits) were highlighted by clinicians as being the most important to treat

The final conceptual model, based on the TLR and VoP report, and refined through feedback from psychiatric clinicians, resulted in a conceptual model with 75 symptoms across 8 domains, and 28 impacts

Figure 2. Final conceptual model based on results from the TLR and the VoP report; refined through qualitative interviews with psychiatric clinicians

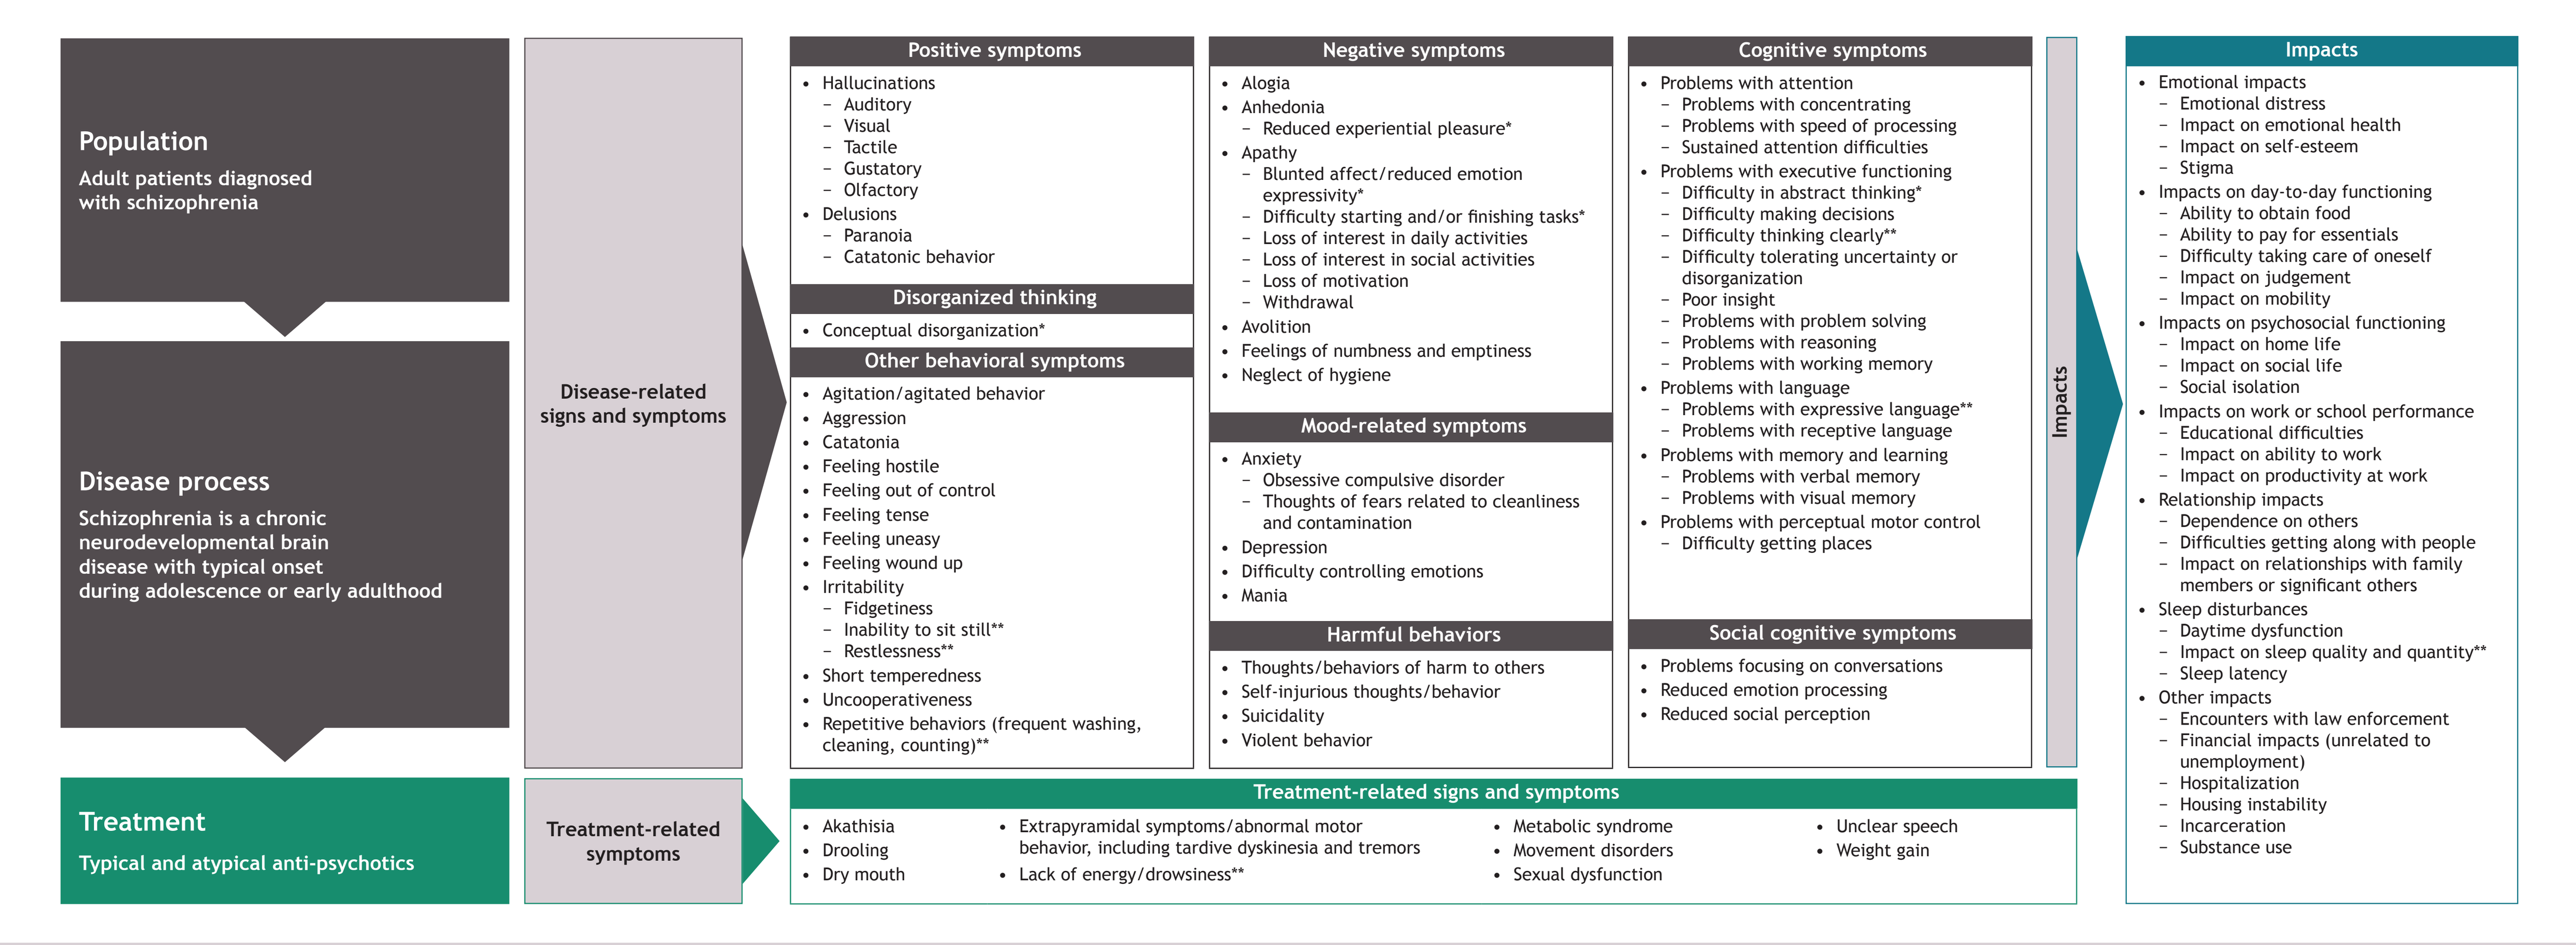
Population
Adult patients diagnosed with schizophrenia
Disease process
Schizophrenia is a chronic neurodevelopmental brain disease with typical onset during adolescence or early adulthood
Treatment
Typical and atypical anti-psychotics
Disease-related signs and symptoms
Treatment-related symptoms
Positive symptoms
• Hallucinations
– Auditory
– Visual
– Tactile
– Gustatory
– Olfactory
• Delusions
– Paranoia
– Catatonic behavior
Disorganized thinking
• Conceptual disorganization*
Other behavioral symptoms
• Agitation/agitated behavior
• Aggression
• Catatonia
• Feeling hostile
• Feeling out of control
• Feeling tense
• Feeling uneasy
• Feeling wound up
• Irritability
– Fidgetiness
– Inability to sit still**
– Restlessness**
• Short temperedness
• Uncooperativeness
• Repetitive behaviors (frequent washing, cleaning, counting)**
Negative symptoms
• Alogia
• Anhedonia
– Reduced experiential pleasure*
• Apathy
– Blunted affect/reduced emotion expressivity*
– Difficulty starting and/or finishing tasks*
– Loss of interest in daily activities
– Loss of interest in social activities
– Loss of motivation
– Withdrawal
• Avolition
• Feelings of numbness and emptiness
• Neglect of hygiene
Mood-related symptoms
• Anxiety
– Obsessive compulsive disorder
– Thoughts of fears related to cleanliness and contamination
• Depression
• Difficulty controlling emotions
• Mania
Harmful behaviors
• Thoughts/behaviors of harm to others
• Self-injurious thoughts/behavior
• Suicidality
• Violent behavior
Cognitive symptoms
• Problems with attention
– Problems with concentrating
– Problems with speed of processing
– Sustained attention difficulties
• Problems with executive functioning
– Difficulty in abstract thinking*
– Difficulty making decisions
– Difficulty thinking clearly**
– Difficulty tolerating uncertainty or disorganization
– Poor insight
– Problems with problem solving
– Problems with reasoning
– Problems with working memory
• Problems with language
– Problems with expressive language**
– Problems with receptive language
• Problems with memory and learning
– Problems with verbal memory
– Problems with visual memory
• Problems with perceptual motor control
– Difficulty getting places
Social cognitive symptoms
• Problems focusing on conversations
• Reduced emotion processing
• Reduced social perception
Treatment-related signs and symptoms
• Akathisia
• Drooling
• Dry mouth
• Extrapyramidal symptoms/abnormal motor behavior, including tardive dyskinesia and tremors
• Lack of energy/drowsiness**
• Metabolic syndrome
• Movement disorders
• Sexual dysfunction
• Unclear speech
• Weight gain
Impacts
Impacts
• Emotional impacts
– Emotional distress
– Impact on emotional health
– Impact on self-esteem
– Stigma
• Impacts on day-to-day functioning
– Ability to obtain food
– Ability to pay for essentials
– Difficulty taking care of oneself
– Impact on judgement
– Impact on mobility
• Impacts on psychosocial functioning
– Impact on home life
– Impact on social life
– Social isolation
• Impacts on work or school performance
– Educational difficulties
– Impact on ability to work
– Impact on productivity at work
• Relationship impacts
– Dependence on others
– Difficulties getting along with people
– Impact on relationships with family members or significant others
• Sleep disturbances
– Daytime dysfunction
– Impact on sleep quality and quantity**
– Sleep latency
• Other impacts
– Encounters with law enforcement
– Financial impacts (unrelated to unemployment)
– Hospitalization
– Housing instability
– Incarceration
– Substance use

*Denotes symptoms that were extracted from articles focusing on treatment-resistant schizophrenia. ** Denotes symptoms that are both disease- and treatment-related.

Table 1. Clinical experience of interviewed psychiatric clinicians

Clinician ID	Years seeing patients with schizophrenia	Number of patients with schizophrenia seen per month	Experience with patients with inadequate response	Clinical setting (inpatient, outpatient, both)	Experience with clinical trials in schizophrenia	Experience with clinical outcome assessments
Clin-01	24	90	Y	Both	Y	Y
Clin-02	17	~100	Y	Both	Y	Y
Clin-03	>30	45–50	Y	Both	Y	Y
Clin-04	6	40	Y	Both	N	Y
Clin-05	19	30	Y	Inpatient	Y	Y
Clin-06	17	300	Y	Both	Y	Y
Clin-07	25	45	Y	Outpatient	Y	Y
Clin-08	20+	Dozens	Y	Both	Y	Y
Clin-09	12	90	Y	Both	Y	Y
Clin-10	13	50	Y	Both	Y	Y

Table 2. Clinician feedback on concepts included in the preliminary conceptual model

Preliminary Conceptual Model Domain	Clinician feedback									
	Clin-01	Clin-02	Clin-03	Clin-04	Clin-05	Clin-06	Clin-07	Clin-08	Clin-09	Clin-10
Positive symptoms										
Remove concepts	No suggestions	No suggestions	At least one suggested change	No suggestions	At least one suggested change	At least one suggested change	At least one suggested change	At least one suggested change	No suggestions	No suggestions
Rename concepts	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change
Reclassify concepts	At least one suggested change	At least one suggested change	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions
Missing concepts	No suggestions	At least one suggested change	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	At least one suggested change
Negative symptoms										
Remove concepts	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Rename concepts	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions
Missing concepts	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions
Cognitive symptoms										
Remove concepts	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions
Rename concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	At least one suggested change
Missing concepts	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Other behavioral symptoms										
Remove concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	Not asked due to time constraints	No suggestions	Not asked due to time constraints
Rename concepts	At least one suggested change	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	Not asked due to time constraints	No suggestions	Not asked due to time constraints
Reclassify concepts	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions	At least one suggested change	Not asked due to time constraints	Not asked due to time constraints	No suggestions	Not asked due to time constraints
Missing concepts	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	Not asked due to time constraints	No suggestions	Not asked due to time constraints
Mood-related symptoms										
Remove concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	Not asked due to time constraints	At least one suggested change	No suggestions	At least one suggested change
Rename concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Missing concepts	At least one suggested change	At least one suggested change	At least one suggested change	No suggestions	At least one suggested change	At least one suggested change	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Harmful behaviors										
Remove concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Rename concepts	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Missing concepts	At least one suggested change	At least one suggested change	No suggestions	No suggestions	No suggestions	At least one suggested change	Not asked due to time constraints	At least one suggested change	At least one suggested change	At least one suggested change
Treatment-related symptoms										
Remove concepts	At least one suggested change	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	At least one suggested change	No suggestions	No suggestions
Rename concepts	At least one suggested change	At least one suggested change	At least one suggested change	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	Not asked due to time constraints	No suggestions	No suggestions	No suggestions
Missing concepts	At least one suggested change	At least one suggested change	At least one suggested change	At least one suggested change	At least one suggested change	At least one suggested change	Not asked due to time constraints	At least one suggested change	At least one suggested change	At least one suggested change
Impacts										
Remove concepts	No suggestions	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Rename concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Reclassify concepts	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions
Missing concepts	No suggestions	At least one suggested change	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	No suggestions	At least one suggested change	No suggestions

No suggestions | At least one suggested change | Not asked due to time constraints

Results (continued)

- Negative and neurocognitive symptoms were specifically noted as areas where current treatments are not sufficient, indicating a significant unmet therapeutic need in schizophrenia care
- Most clinicians (n=8; 2 clinicians were not asked due to time constraints) reported that antipsychotic medications were their primary choice of treatment, with efficacy and safety driving their choice of treatment

Refinement of the conceptual model

- The preliminary conceptual model developed through the TLR was considered generally comprehensive by clinicians
- Based on clinician feedback, 7 concepts were removed from the preliminary conceptual model, 14 concepts were renamed or reclassified under different domains (**Table 2**), and 15 new concepts were added
- The final conceptual model included 75 symptoms across 8 domains (positive symptoms, negative symptoms, cognitive symptoms, disorganized thinking, other behavioral symptoms, mood-related symptoms, harmful behaviors, and social cognitive symptoms) and 28 impacts associated with these domains (**Figure 2**)

Conclusions

- Insights from the VoP and TLR revealed a broad range of signs, symptoms, and impacts experienced by PLS, underscoring the significant burden on daily life
- These findings informed the development of a preliminary conceptual model designed to assess psychosocial functioning and visually represent symptom impact
- Qualitative interviews with psychiatrists confirmed the disease and treatment burden identified in the literature and highlighted a substantial unmet treatment need in PLS
- Clinician feedback led to refinement of the conceptual model, which can serve as a foundation for clinical outcome assessments, research, and as a tool to enhance clinician understanding of schizophrenia's impact on daily life

References

1. Goldstone LW. Am J Manag Care. 2020;26(3 Suppl):S48–S54.
2. Kantrowitz JT, et al. Int J Neuropsychopharmacol. 2023;26(5):322–330.
3. Powers JH III, et al. Value Health. 2017;20(1):2–4.
4. American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders. 5th ed. American Psychiatric Association; 2013.
5. Schizophrenia & Psychosis Action Alliance. Reimagine Schizophrenia: Transforming How We Are Treated, Function and Thrive. 2023. Available at: https://sczaction.org/wp-content/uploads/2023/02/Reimagine-Schizophrenia-VOP-report-FINAL-1.pdf.

Acknowledgments

All authors contributed to and approved the poster. This analysis was supported by Bristol Myers Squibb. Medical writing and editorial assistance were provided by IQVIA, funded by Bristol Myers Squibb.

Declaration of interests

This study was funded by Bristol Myers Squibb. SK, JP, BF, and AK are employees of IQVIA, which received funding from Bristol Myers Squibb to conduct this study. SD, and KG are employees of Bristol Myers Squibb, which funded this study. AC was an employee of Bristol Myers Squibb at the time the analysis was conducted. CG is president and principal consultant at Gwaltney Consulting, which received funding from Bristol Myers Squibb to conduct part of this research.

Scientific Content on Demand

QR codes are valid for 30 days after the congress presentation date.

Presented at the Psych Congress, September 17–21, 2025, San Diego, CA, US

Email: Kristin.Gillard@bms.com

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.